Serial circulating tumor DNA (ctDNA) monitoring in early-stage, HR+/HER2-, invasive lobular carcinoma (ILC) of the breast and impact on clinical outcomes

Julia Foldi,[1] Samuel Rivero-Hinojosa,[2] Sandro Satta,[2] Sara L. Bristow,[2] Ekaterina Kalashnikova,[2] Steffi Oesterreich,[1] Adrian V. Lee,[1] Marija Balic,[1] Angel A. Rodriguez,[2] Minetta C Liu[2]

[1]UPMC Hillman Cancer Center, Magee-Womens Research Institute, University of Pittsburgh and UPMC, Pittsburgh, PA, USA; [2]Natera, Inc., Austin, TX, USA

Background

- ILC accounts for ~10-15% of breast cancer cases in the US, with difficult-to-diagnose patterns of metastasis.
- Loss of E-cadherin (*CDH1*) is the pathognomonic feature of ILC.
- Tumor-informed ctDNA assays detect molecular residual disease (MRD) in patients who completed definitive therapy for early-stage breast cancer, however, little is known about the ctDNA dynamics and their prognostic relevance in ILC specifically.
- Here, we assessed genomic alterations and correlated ctDNA dynamics with clinical outcomes in patients with HR+/HER2- ILC.

Methods

- We utilized Natera's proprietary real-world database, which is linked to commercially available claims data and results of commercial ctDNA testing using a clinically validated, personalized, tumor-informed mPCR-NGS ctDNA assay (Signatera™, Natera, Inc.), to identify patients with early-stage breast cancer.
- Lobular, ductal, or mixed histologic subtype was established based on clinical notes and pathology reports.
- Tumor subtypes defined by ER and HER2 status were categorized using treatment regimens from insurance claim codes.
- Date of distant relapse was obtained from secondary malignant neoplasm claims codes.
- ctDNA positivity rates and distant recurrence-free survival (DRFS) were evaluated using longitudinal ctDNA status (ctDNA-positive or -negative).
- The timing of ctDNA positivity after primary breast cancer surgery was divided into two categories: <2 years and >2 years.

Figure 1. CONSORT diagram

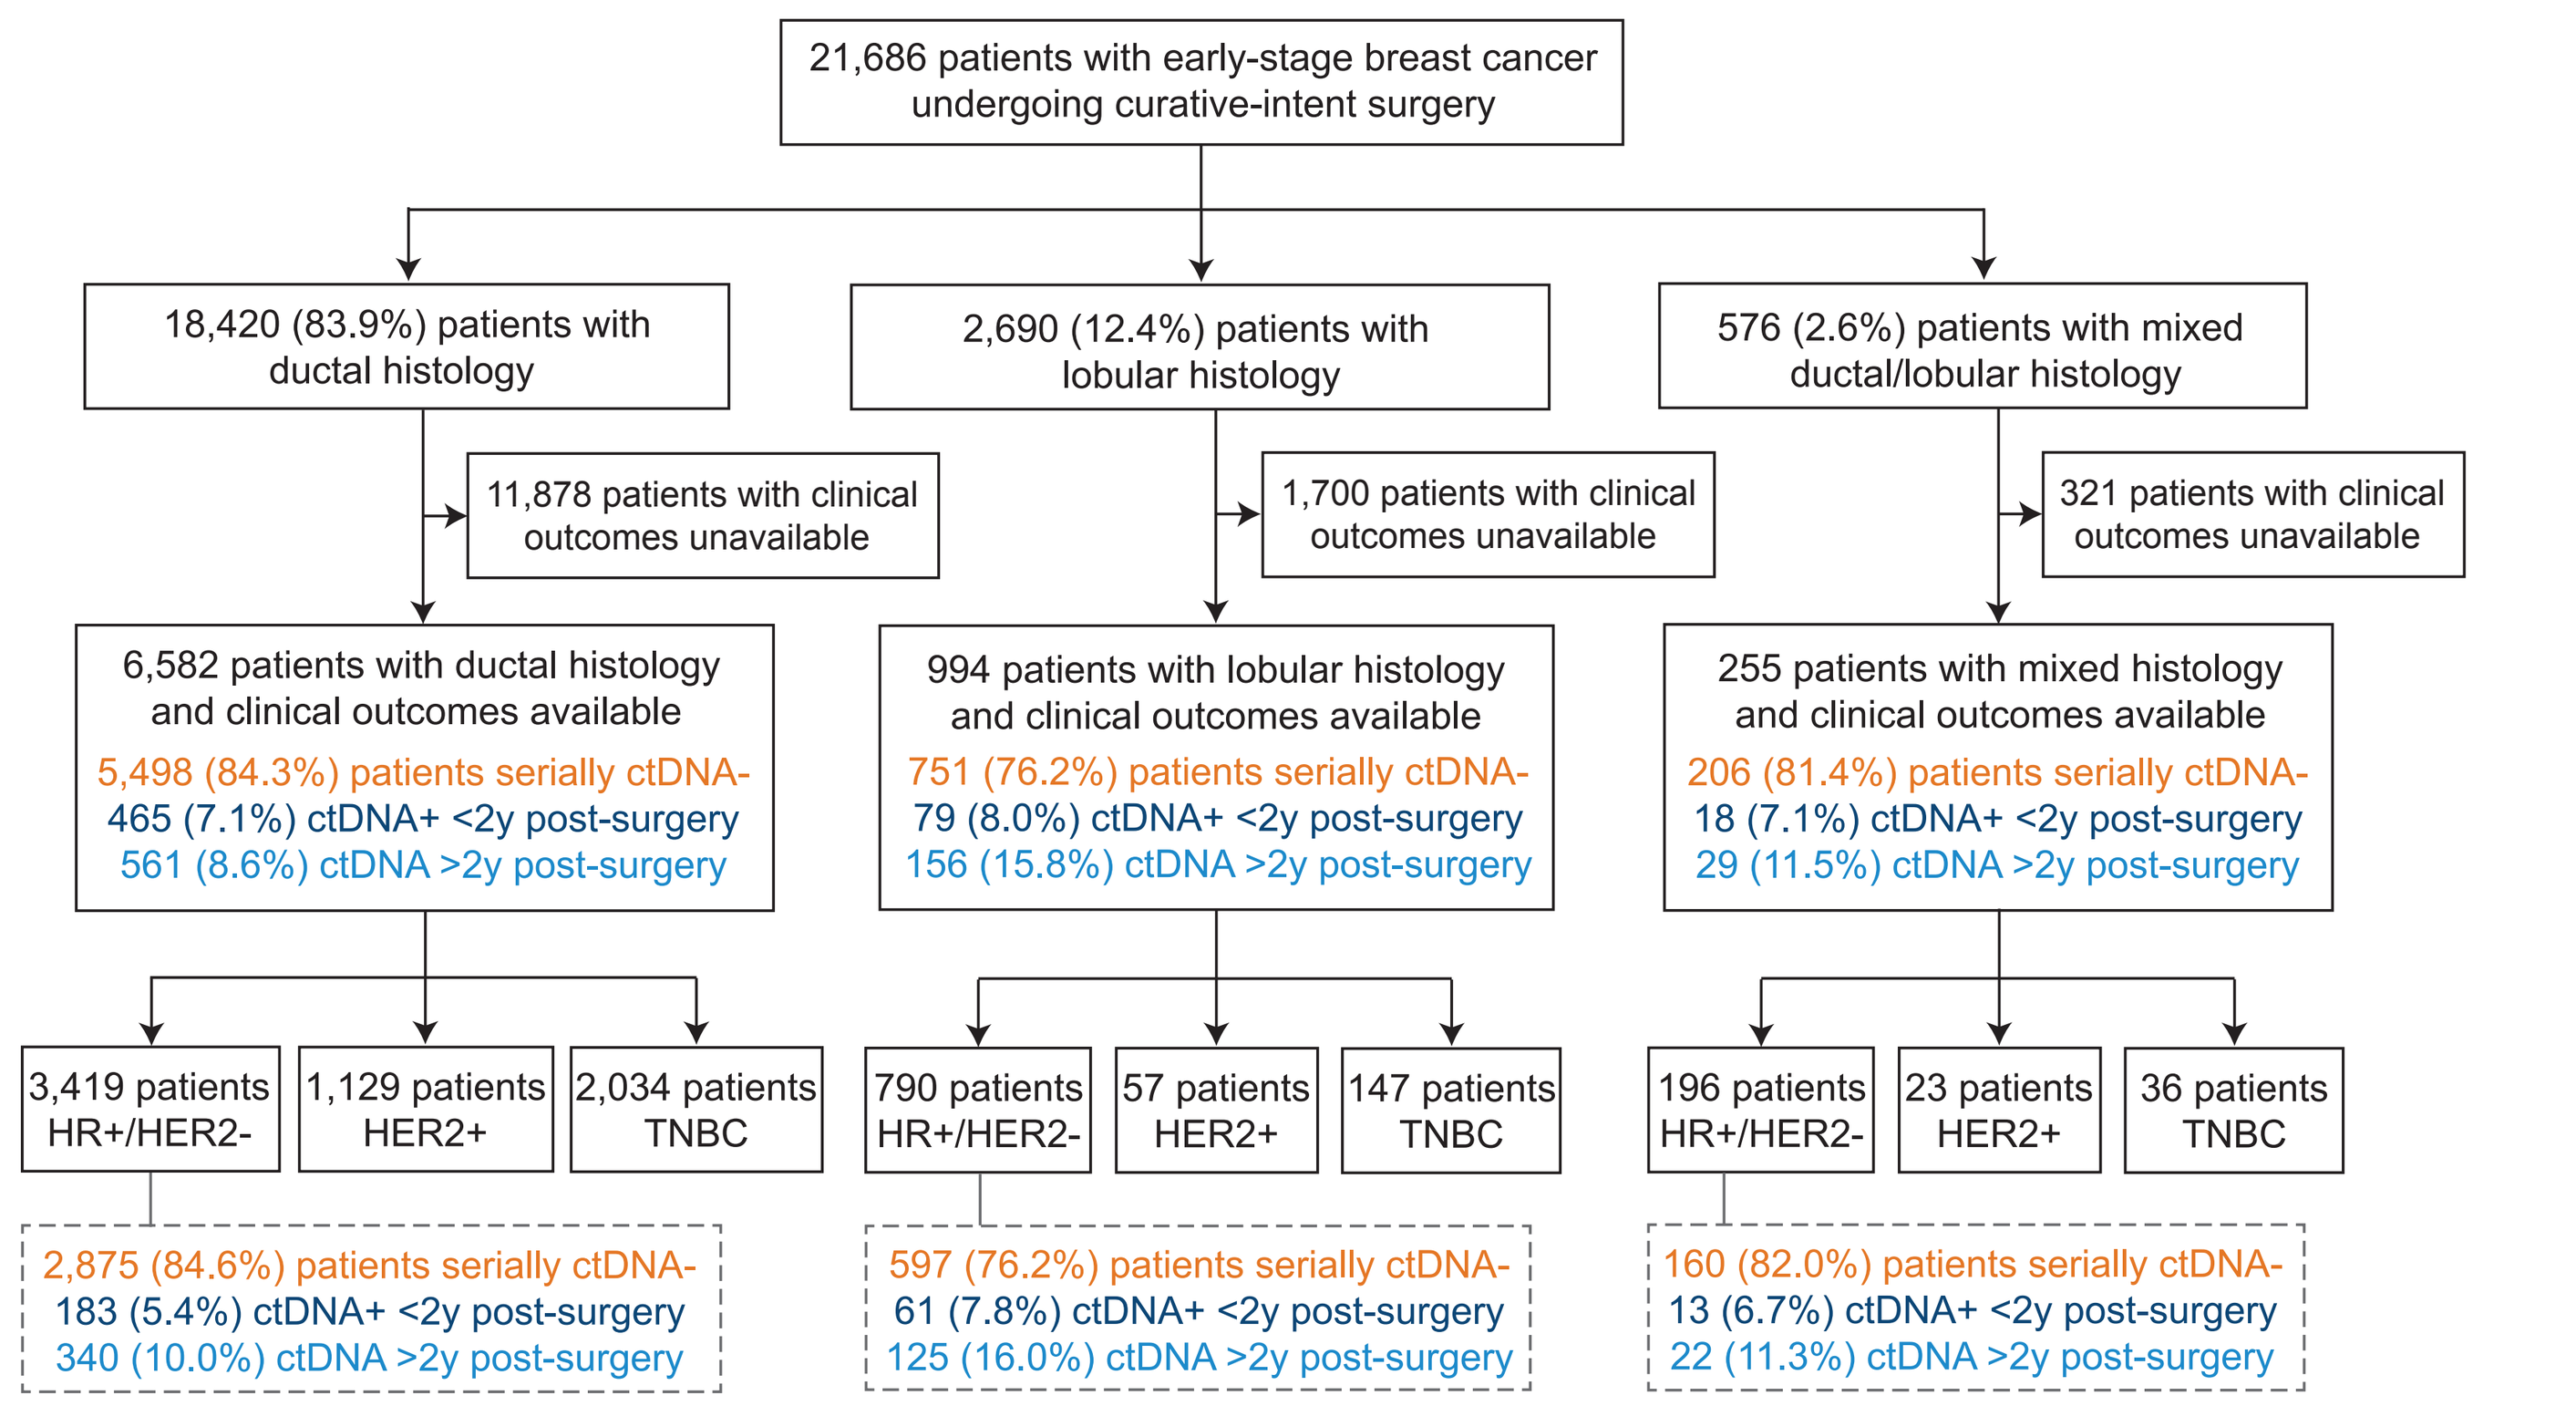

21,686 patients with early-stage breast cancer undergoing curative-intent surgery
18,420 (83.9%) patients with ductal histology
2,690 (12.4%) patients with lobular histology
576 (2.6%) patients with mixed ductal/lobular histology
11,878 patients with clinical outcomes unavailable
1,700 patients with clinical outcomes unavailable
321 patients with clinical outcomes unavailable
6,582 patients with ductal histology and clinical outcomes available
5,498 (84.3%) patients serially ctDNA-
465 (7.1%) ctDNA+ <2y post-surgery
561 (8.6%) ctDNA >2y post-surgery
994 patients with lobular histology and clinical outcomes available
751 (76.2%) patients serially ctDNA-
79 (8.0%) ctDNA+ <2y post-surgery
156 (15.8%) ctDNA >2y post-surgery
255 patients with mixed histology and clinical outcomes available
206 (81.4%) patients serially ctDNA-
18 (7.1%) ctDNA+ <2y post-surgery
29 (11.5%) ctDNA >2y post-surgery
3,419 patients HR+/HER2-
1,129 patients HER2+
2,034 patients TNBC
790 patients HR+/HER2-
57 patients HER2+
147 patients TNBC
196 patients HR+/HER2-
23 patients HER2+
36 patients TNBC
2,875 (84.6%) patients serially ctDNA-
183 (5.4%) ctDNA+ <2y post-surgery
340 (10.0%) ctDNA >2y post-surgery
597 (76.2%) patients serially ctDNA-
61 (7.8%) ctDNA+ <2y post-surgery
125 (16.0%) ctDNA >2y post-surgery
160 (82.0%) patients serially ctDNA-
13 (6.7%) ctDNA+ <2y post-surgery
22 (11.3%) ctDNA >2y post-surgery

Figure 1. Flow chart depicting the breakdown of the cohort by histologic and hormone subtypes, along with ctDNA-positivity rates. Abbreviations: TNBC, triple-negative breast cancer; ctDNA+, ctDNA-positive; ctDNA-, ctDNA-negative; y, years.

Conclusions

- To our knowledge, this is the first large-scale analysis of the landscape of targetable genomic alterations and matching longitudinal ctDNA results and outcomes in patients with early-stage breast cancer.
- In patients with lobular histology, *PIK3CA* is most frequently co-mutated with *CDH1*.
- Patients with lobular phenotype (HR+/HER2-) demonstrated longer time to recurrence and longitudinally higher rates of ctDNA positivity compared to ductal and mixed phenotypes.
- Longitudinally, ctDNA status after surgery is predictive of outcomes regardless of the subtype.

Acknowledgments & Disclosures

S.R.-H., S.S., S.B., E.K.., A.A.R, and M.C.L. are employees at Natera, Inc. with stock or options to own stock.

Patients with HR+/HER2- ILC demonstrated longitudinally higher rates of ctDNA positivity and longer time to relapse compared to those with ductal and mixed lobular/ductal subtypes.

Figure 2. Genes most frequently mutated in patients with lobular subtype compared to ductal and mixed subtypes

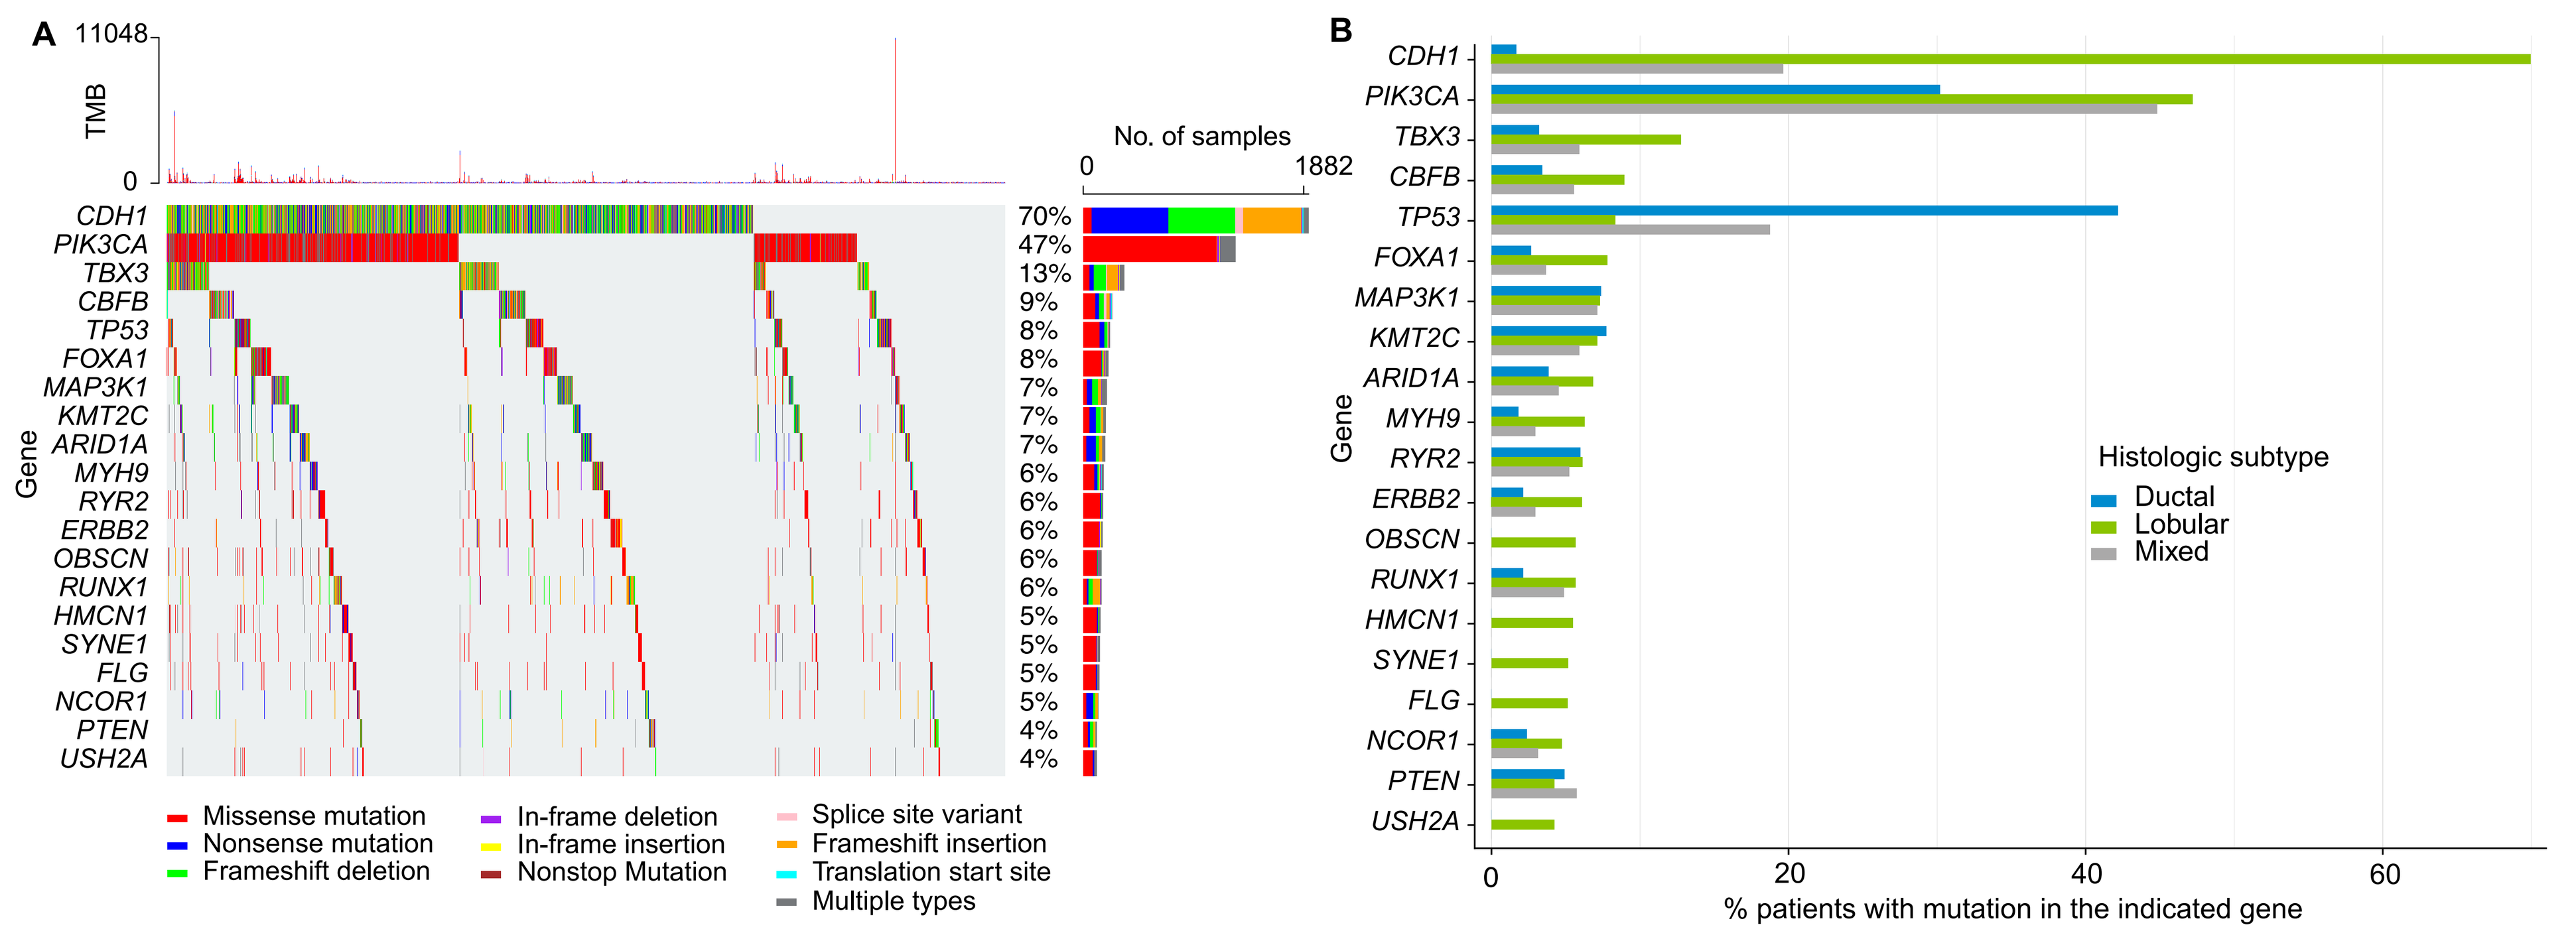

A
11048
TMB
0
No. of samples
0
1882
Gene
CDH1 70%
PIK3CA 47%
TBX3 13%
CBFB 9%
TP53 8%
FOXA1 8%
MAP3K1 7%
KMT2C 7%
ARID1A 7%
MYH9 6%
RYR2 6%
ERBB2 6%
OBSCN 6%
RUNX1 6%
HMCN1 5%
SYNE1 5%
FLG 5%
NCOR1 5%
PTEN 4%
USH2A 4%
Missense mutation
Nonsense mutation
Frameshift deletion
In-frame deletion
In-frame insertion
Nonstop Mutation
Splice site variant
Frameshift insertion
Translation start site
Multiple types
B
Histologic subtype
Ductal
Lobular
Mixed
0
20
40
60
% patients with mutation in the indicated gene

Figure 2. **A.** Among 2,690 patients with ILC, alterations in at least one gene were observed in 2,481 (92.2%) of the patients. The oncoplot depicts the top 20 most frequently mutated genes in patients with ILC. **B.** The bar plot shows the relative frequencies of these 20 genes in patients with lobular, ductal and mixed subtypes.

Figure 3. Cumulative ctDNA positivity rate, rate of relapse, and time to relapse by histologic subtypes

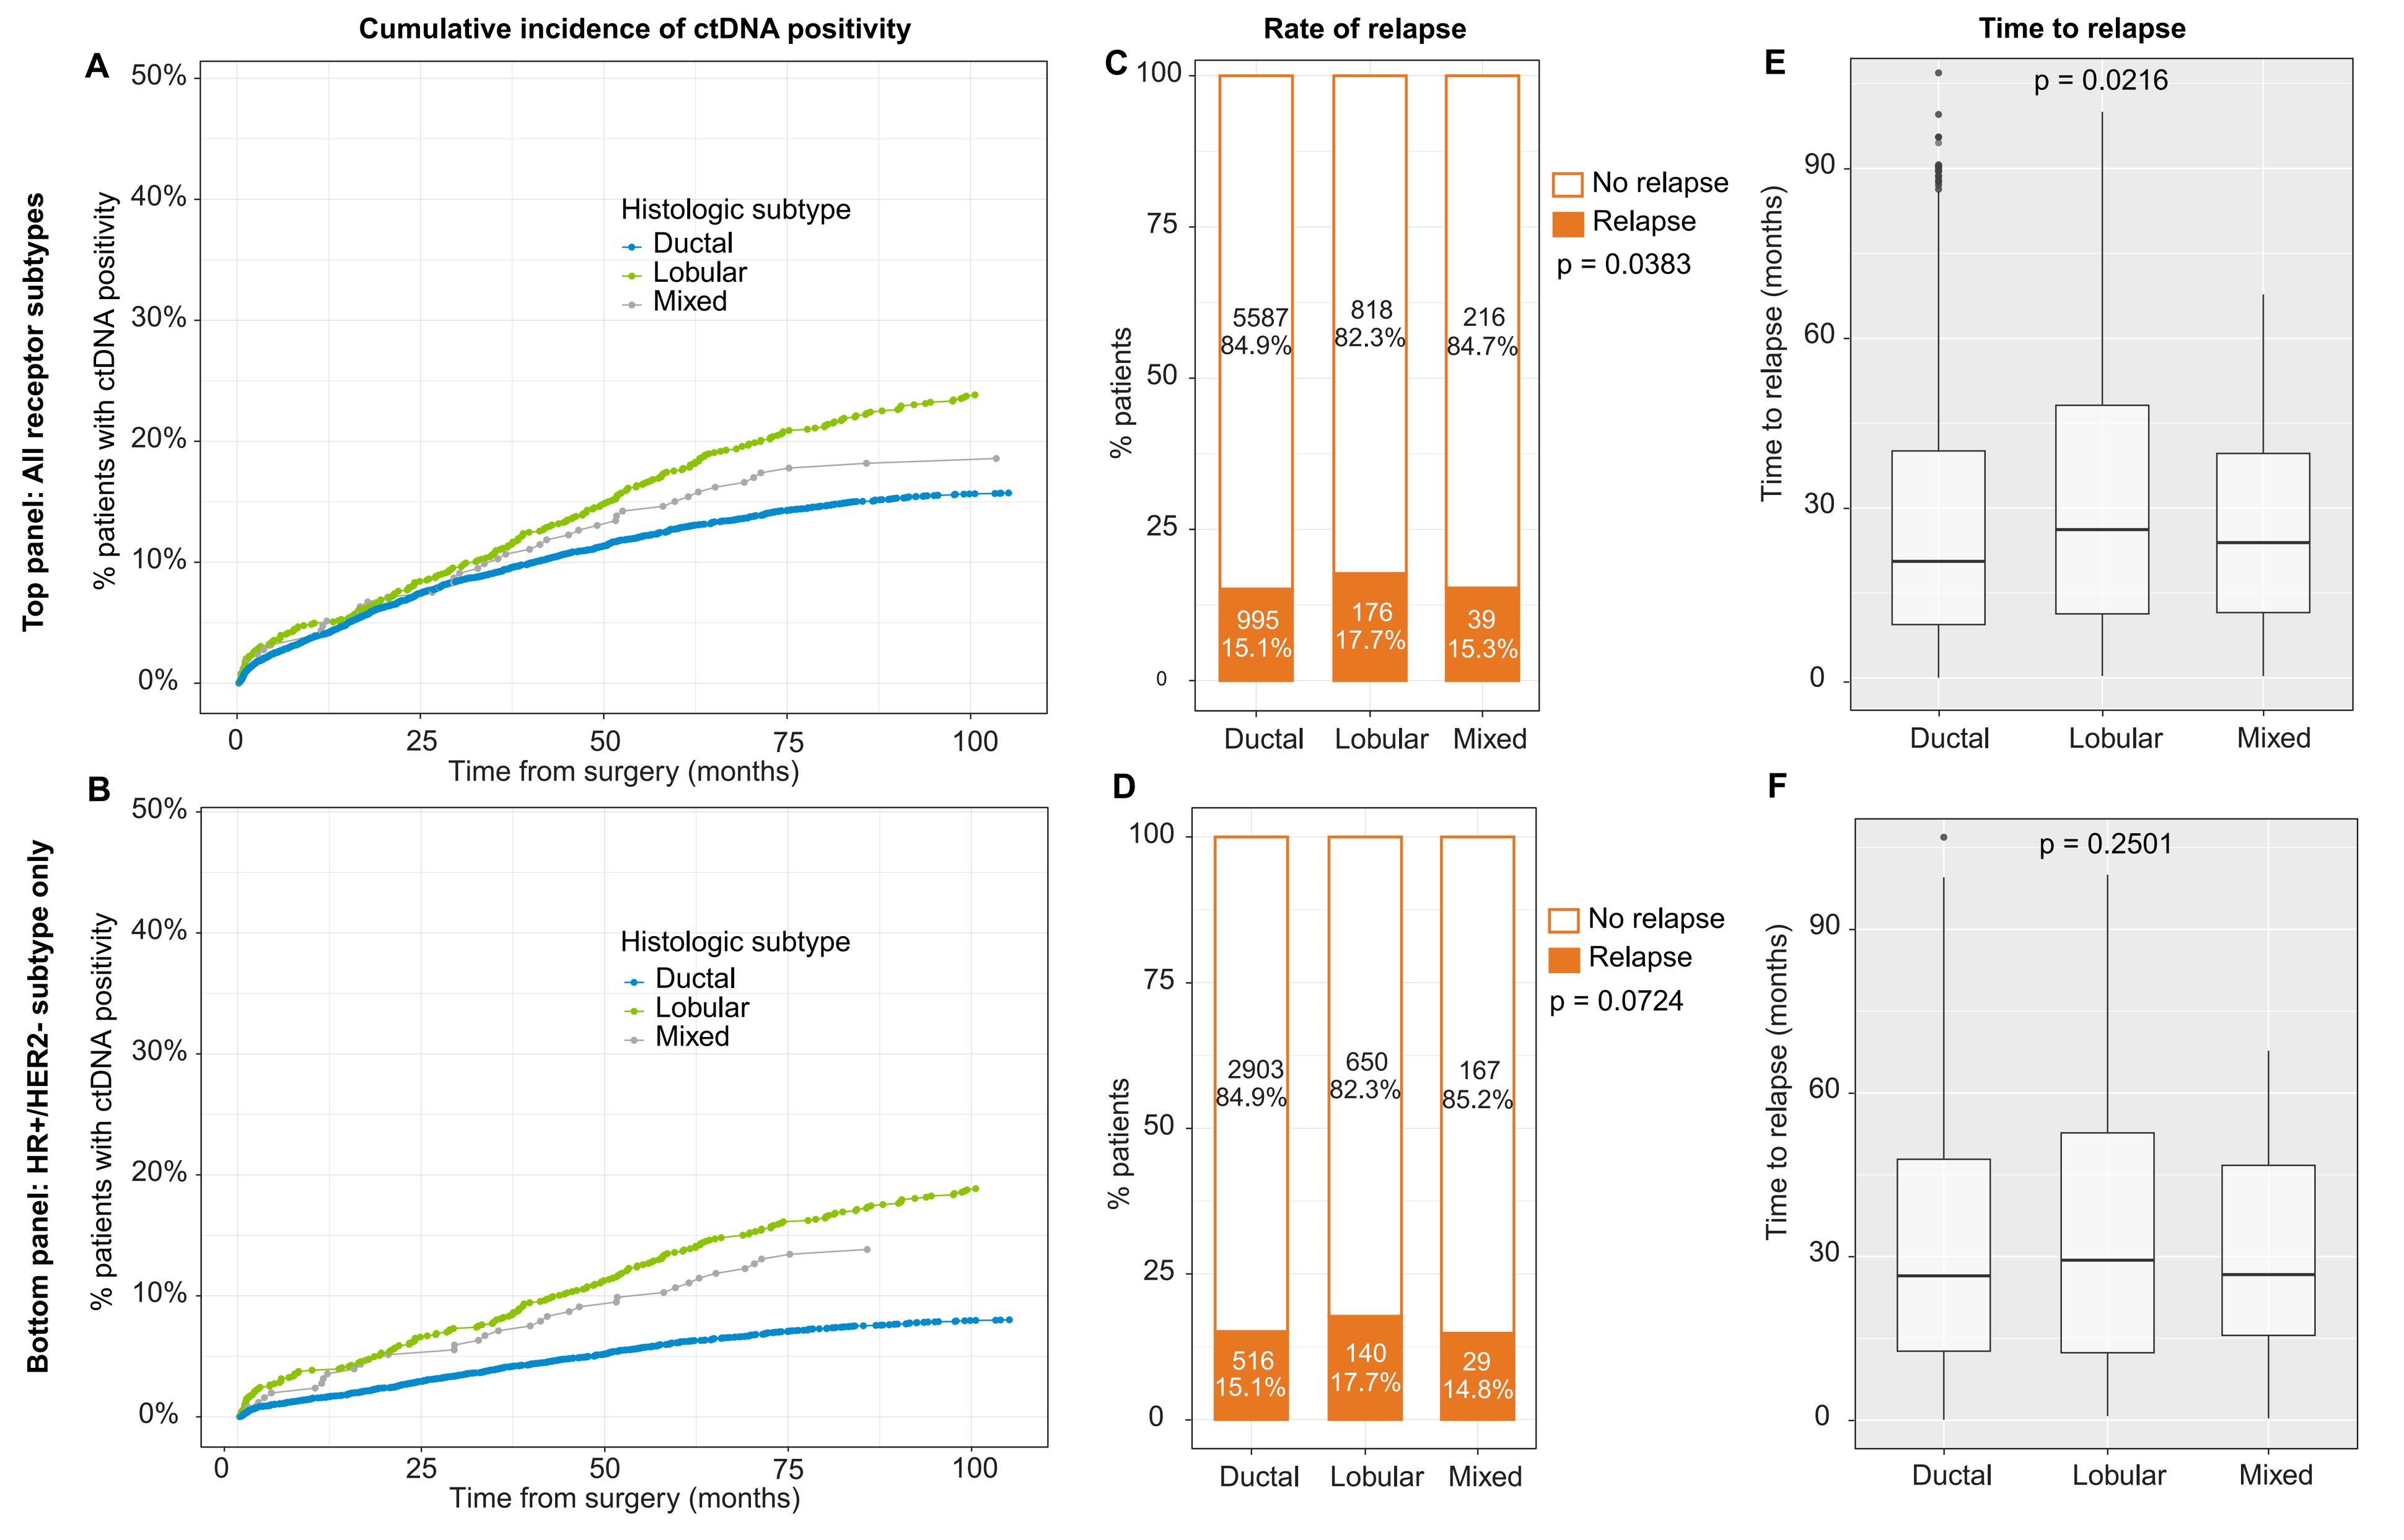

Cumulative incidence of ctDNA positivity
Rate of relapse
Time to relapse
Top panel: All receptor subtypes
Bottom panel: HR+/HER2- subtype only
A
B
% patients with ctDNA positivity
Histologic subtype
Ductal
Lobular
Mixed
Time from surgery (months)
C
D
% patients
5587
84.9%
818
82.3%
216
84.7%
995
15.1%
176
17.7%
39
15.3%
No relapse
Relapse
p = 0.0383
2903
84.9%
650
82.3%
167
85.2%
516
15.1%
140
17.7%
29
14.8%
p = 0.0724
Ductal
Lobular
Mixed
E
F
Time to relapse (months)
p = 0.0216
p = 0.2501

Figure 3. Among 7,831 patients with clinical outcomes available, lobular phenotype was associated with higher cumulative MRD rate (A-B), elevated observed relapse rate (C-D) and increased time to relapse from surgery (E-F) when compared to ductal and mixed histology.

Figure 4. Association of ctDNA status post surgery with DRFS

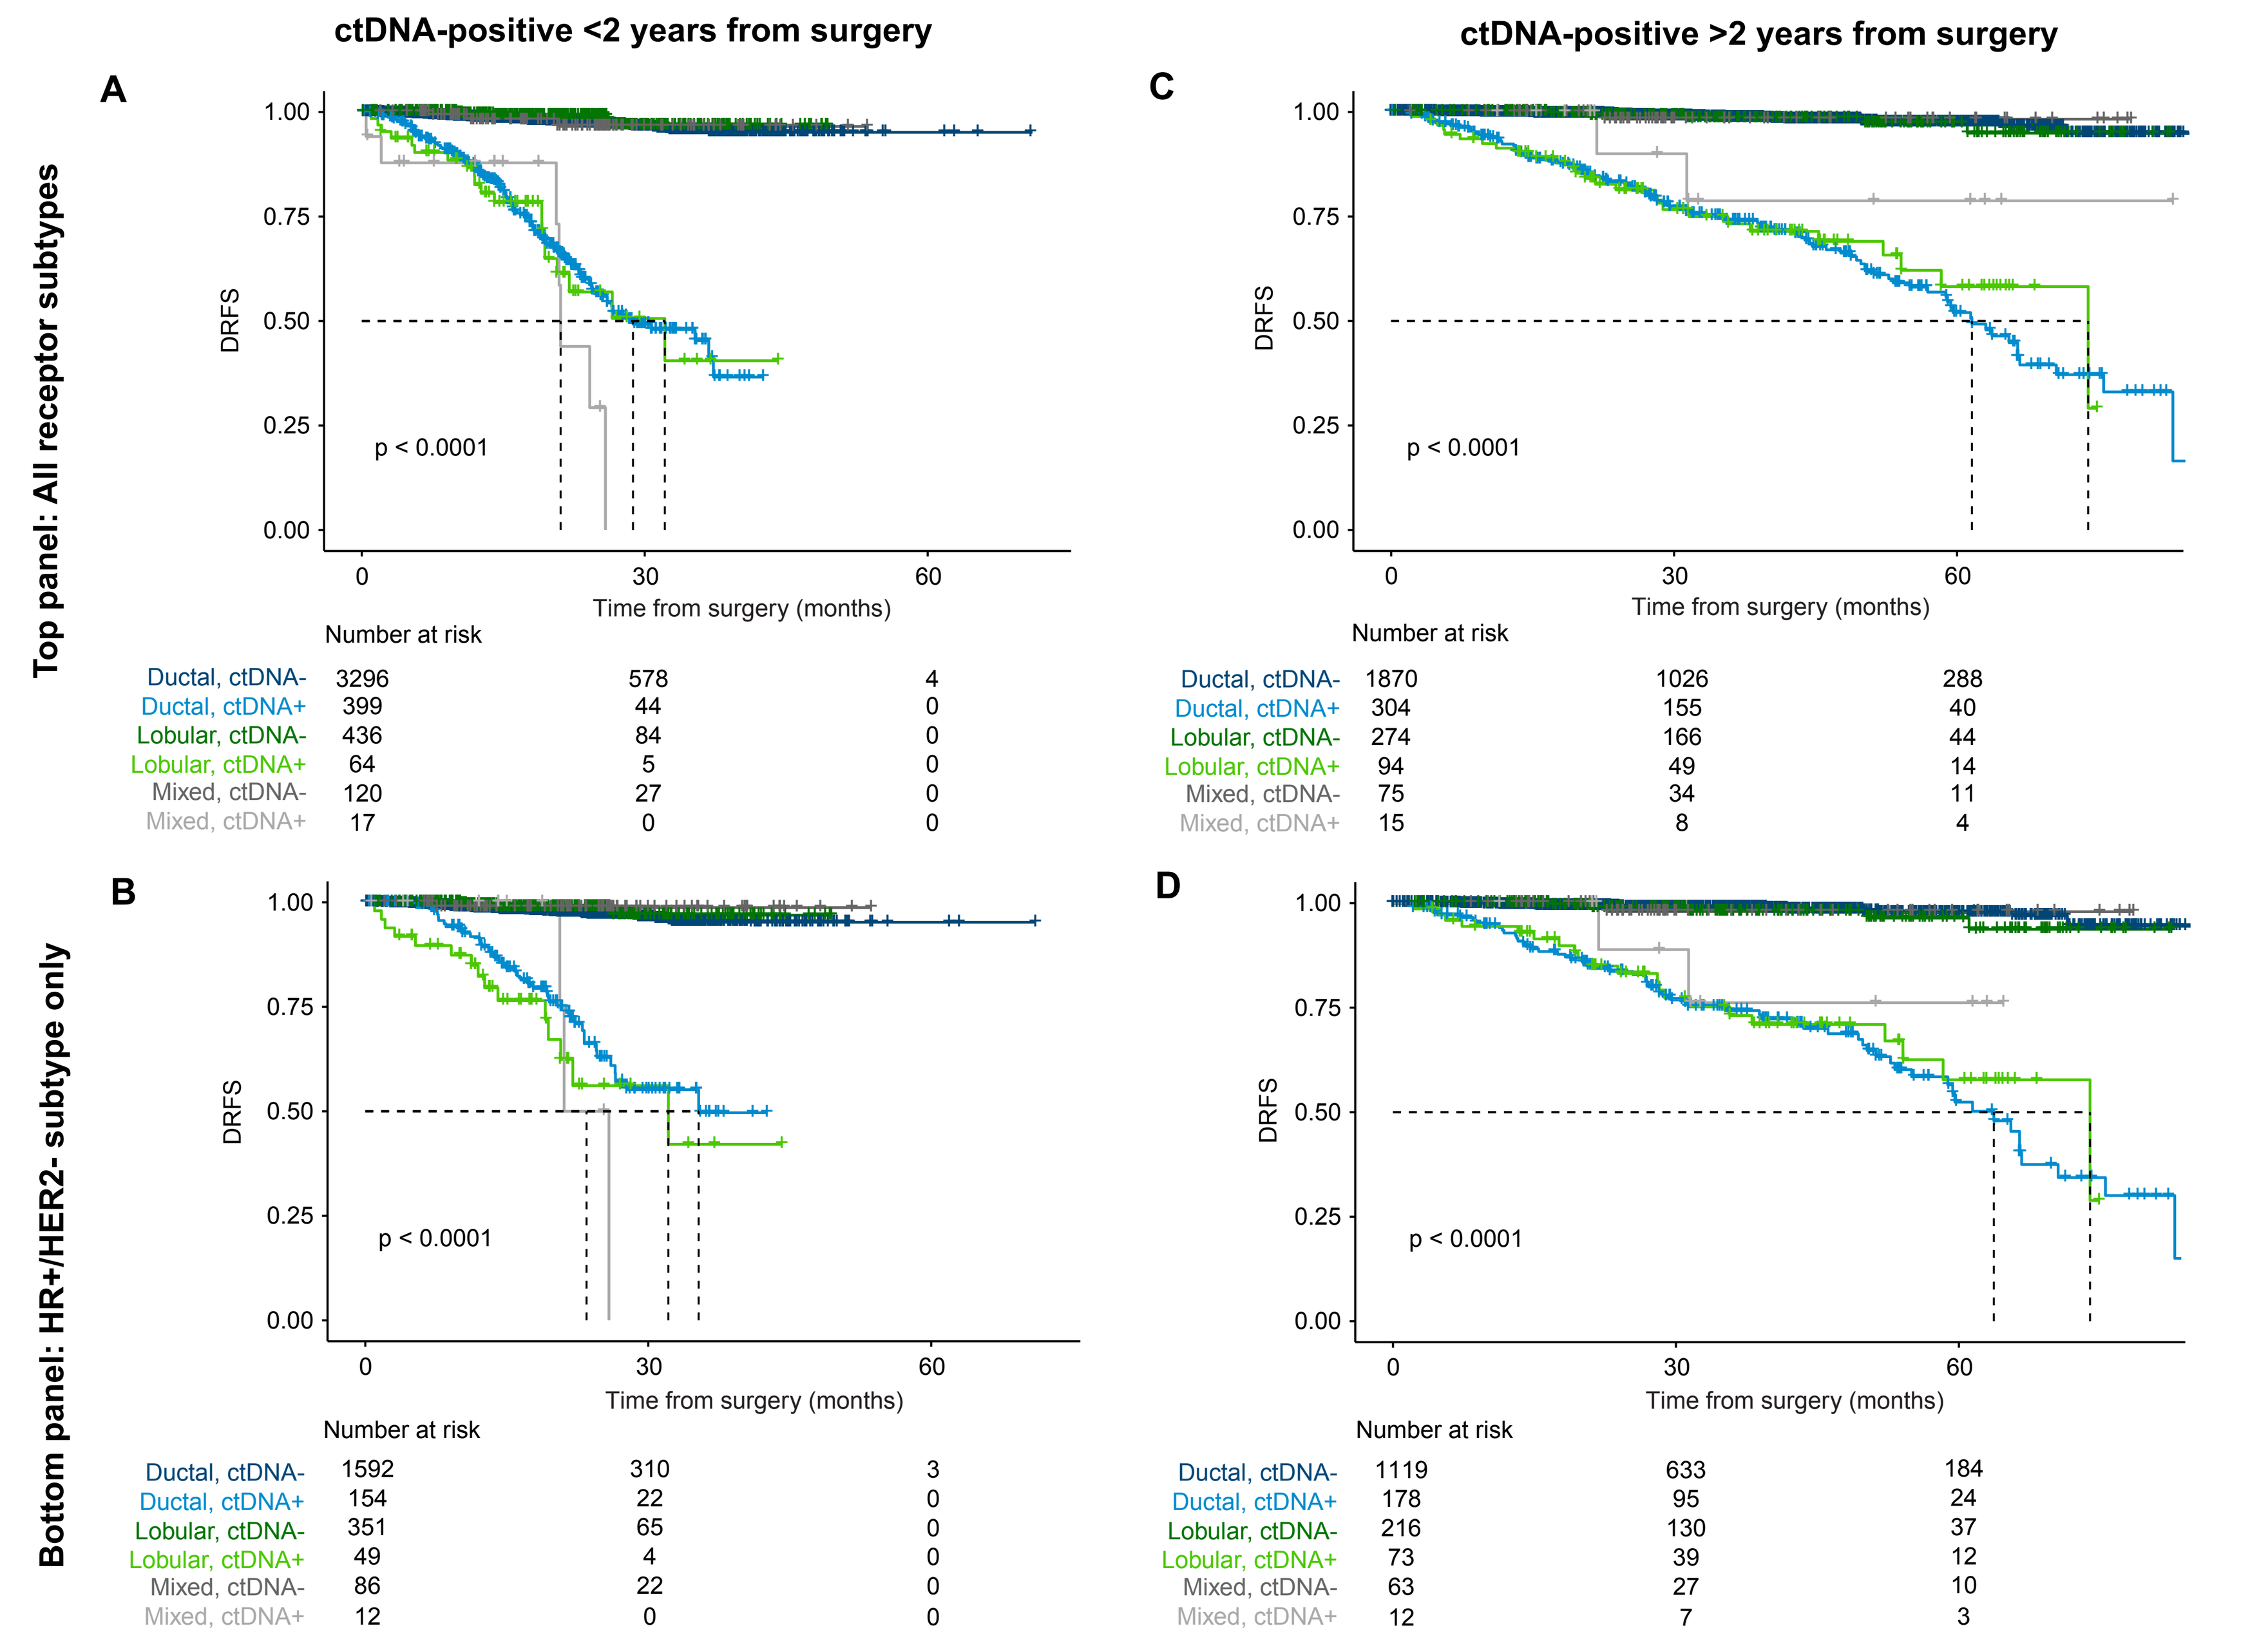

ctDNA-positive <2 years from surgery
ctDNA-positive >2 years from surgery
Top panel: All receptor subtypes
Bottom panel: HR+/HER2- subtype only
A
B
C
D
DRFS
p < 0.0001
Time from surgery (months)
Number at risk
Ductal, ctDNA- 3296 578 4
Ductal, ctDNA+ 399 44 0
Lobular, ctDNA- 436 84 0
Lobular, ctDNA+ 64 5 0
Mixed, ctDNA- 120 27 0
Mixed, ctDNA+ 17 0 0
Ductal, ctDNA- 1870 1026 288
Ductal, ctDNA+ 304 155 40
Lobular, ctDNA- 274 166 44
Lobular, ctDNA+ 94 49 14
Mixed, ctDNA- 75 34 11
Mixed, ctDNA+ 15 8 4
Ductal, ctDNA- 1592 310 3
Ductal, ctDNA+ 154 22 0
Lobular, ctDNA- 351 65 0
Lobular, ctDNA+ 49 4 0
Mixed, ctDNA- 86 22 0
Mixed, ctDNA+ 12 0 0
Ductal, ctDNA- 1119 633 184
Ductal, ctDNA+ 178 95 24
Lobular, ctDNA- 216 130 37
Lobular, ctDNA+ 73 39 12
Mixed, ctDNA- 63 27 10
Mixed, ctDNA+ 12 7 3

Figure 4. **A-B. Early ctDNA positivity:** Among patients of all breast cancer subtypes (A) and HR+/HER2- breast cancer (B), stratification by ctDNA status within two years of surgery identified patient populations with aggressive disease and significantly inferior DRFS**. C-D. Late ctDNA positivity:** Among patients of all repetor subtypes (A) and HR+/HER2- breast cancer (B), stratification by ctDNA status any time after >2 years from surgery identified patient populations with late recurrence. ctDNA-, ctDNA-negative; ctDNA+, ctDNA-positive; DRFS, distant recurrence-free survival.

Figure 5. Patients with lobular subtype harboring *CDH1* mutation have higher cumulative ctDNA-positivity rate

A

Subtype	Gene	N	# patients with mutation (%)
Ductal	*CDH1*	18,423	306 (1.7)
Ductal	*PIK3CA*	18,423	5,552 (30.1)
Lobular	*CDH1*	2,694	1,882 (69.9)
Lobular	*PIK3CA*	2,694	1,270 (47.1)
Mixed	*CDH1*	576	113 (19.6)
Mixed	*PIK3CA*	576	258 (44.8)

B

Subtype	mut *CDH1* mut *PIK3CA*	mut *CDH1* wt *PIK3CA*	wt *CDH1* mut *PIK3CA*	wt *CDH1* wt *PIK3CA*
Ductal	0.8	0.9	29.4	69.0
Lobular	34.8	35.1	12.4	17.8
Mixed	12.3	7.3	32.5	47.9

C

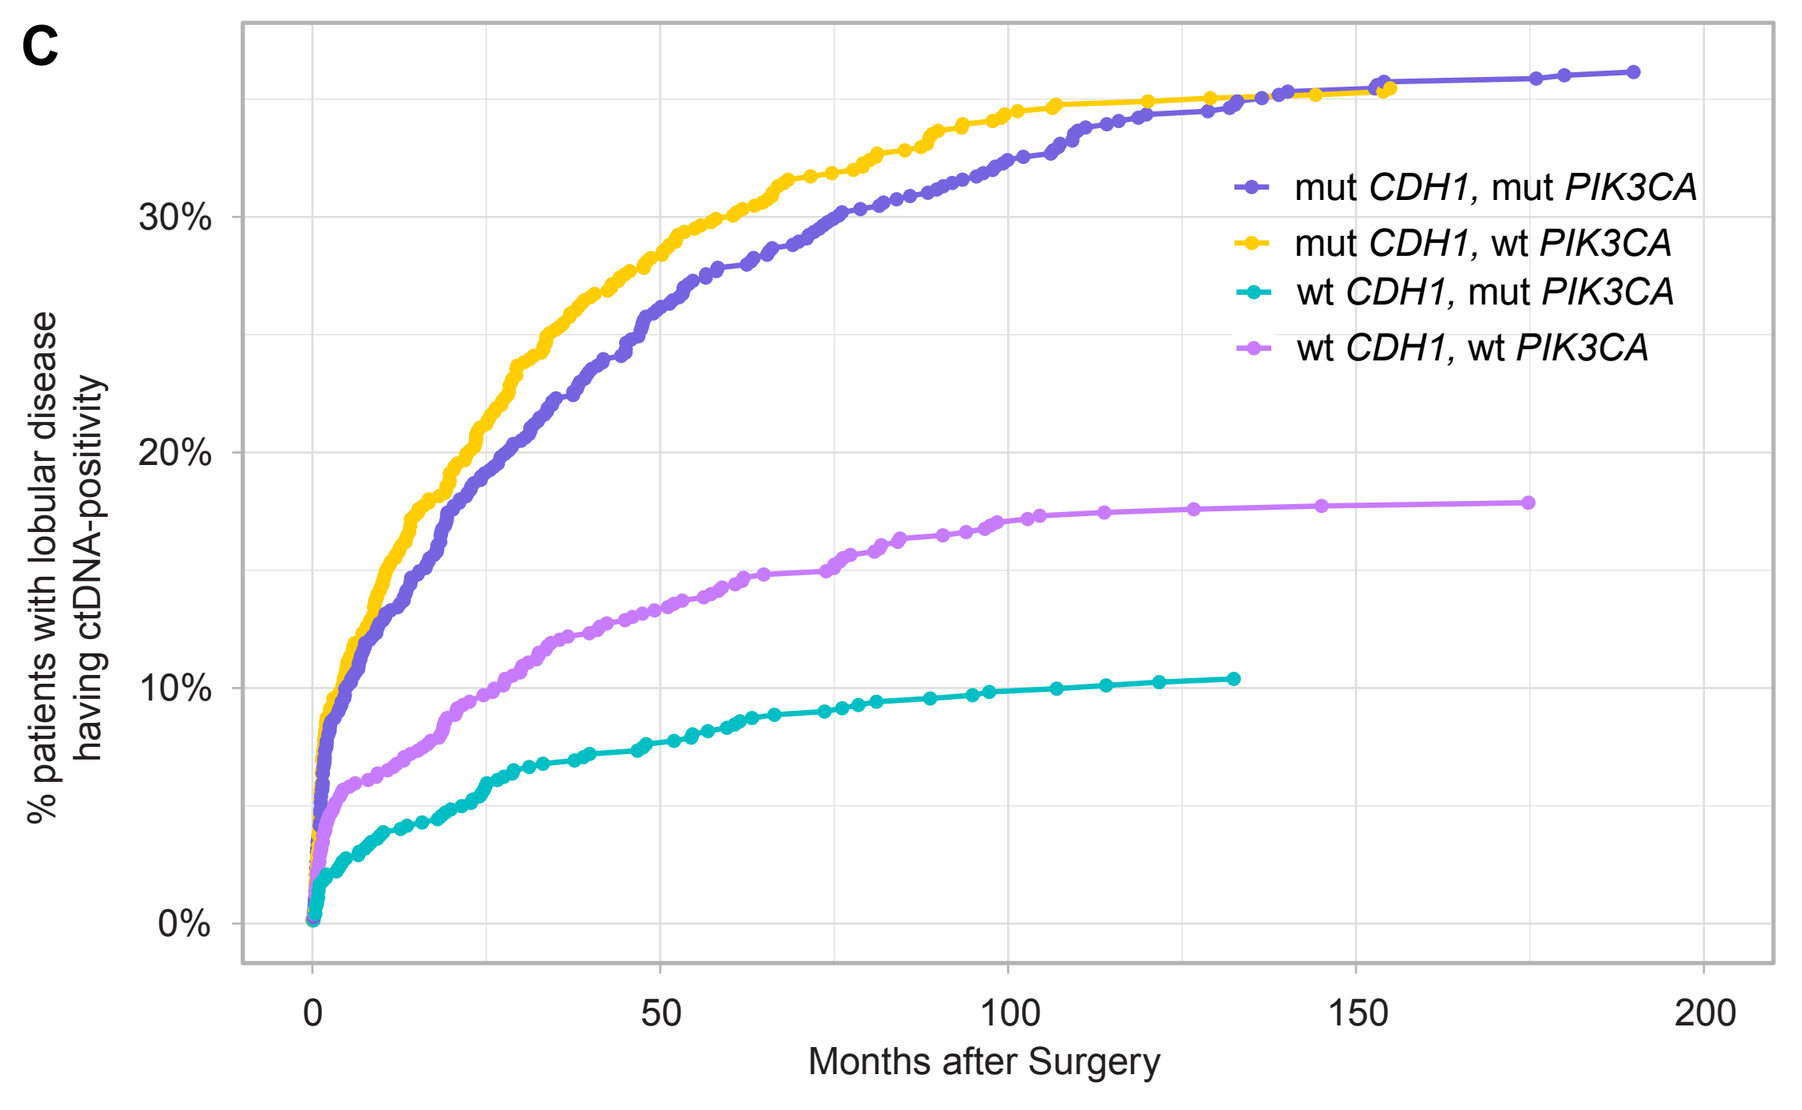

% patients with lobular disease having ctDNA-positivity
0%
10%
20%
30%
mut CDH1, mut PIK3CA
mut CDH1, wt PIK3CA
wt CDH1, mut PIK3CA
wt CDH1, wt PIK3CA
0
50
100
150
200
Months after Surgery

Figure 5. A. Incidence of *CDH1* and *PIK3CA* mutations observed in the cohort by histologic subtype. **B.** Percent of patients with *CDH1* and *PIK3CA* co-mutations by morphologic subtype. **C.** Cumulative incidence plot depicting the timeline of patients with lobular breast cancer having *CDH1* and/or *PI3K* mutations turning ctDNA-positive after surgery. Elevated cumulative ctDNA positivity rate was observed among those harboring *CDH1* mutations irrespective of *PIK3CA* mutation status. Significantly lower cumulative ctDNA positivity rates are observed in patients with lobular subtype having wt *CDH1*. Abbreviations: wt, wildtype; mut, mutant.

American Society of Clinical Oncology | 2025 ASCO Annual Meeting | May 30 - June 3, 2025 | Chicago, IL | Abstract ID: 581